Streamlined QbD HPLC Method Development with Automated Data Processing

Gang Xue[1], Jeff Harwood[1], Charles Cheng[1], David Fortin[1], Jian Wang[1], George Reid[1], Mike McBrien[2], and Andrey Vazhentsev[2]

[1]Analytical Development, Pfizer Global R&D, Groton, CT 06340

[2]Advanced Chemistry Development, Inc., 110 Yonge Street, 14th Floor, Toronto, Ontario Canada M5C 1T4

Introduction

Quality by Design (QbD) has been widely applied to pharmaceutical manufacturing to enhance process robustness, and improve product quality and manufacturing productivity. Recently, there arose tremendous interests from both the pharmaceutical industry and regulatory agents to expand it to analytical methodology. Similar to process QbD, analytical QbD offers the benefits of more thorough understanding of both the measurement requirements and analytical methods, thus delivers more robust methods. In addition, analytical QbD could significantly reduce the efforts relating to post approval variation. However, the approach would also mean significantly more experimentation to assess and understand the analytical method. This paper describes our attempts to apply some informatics tools to streamline the QbD method development to alleviate the increased resource burden.

Quality by Design Principles[1]

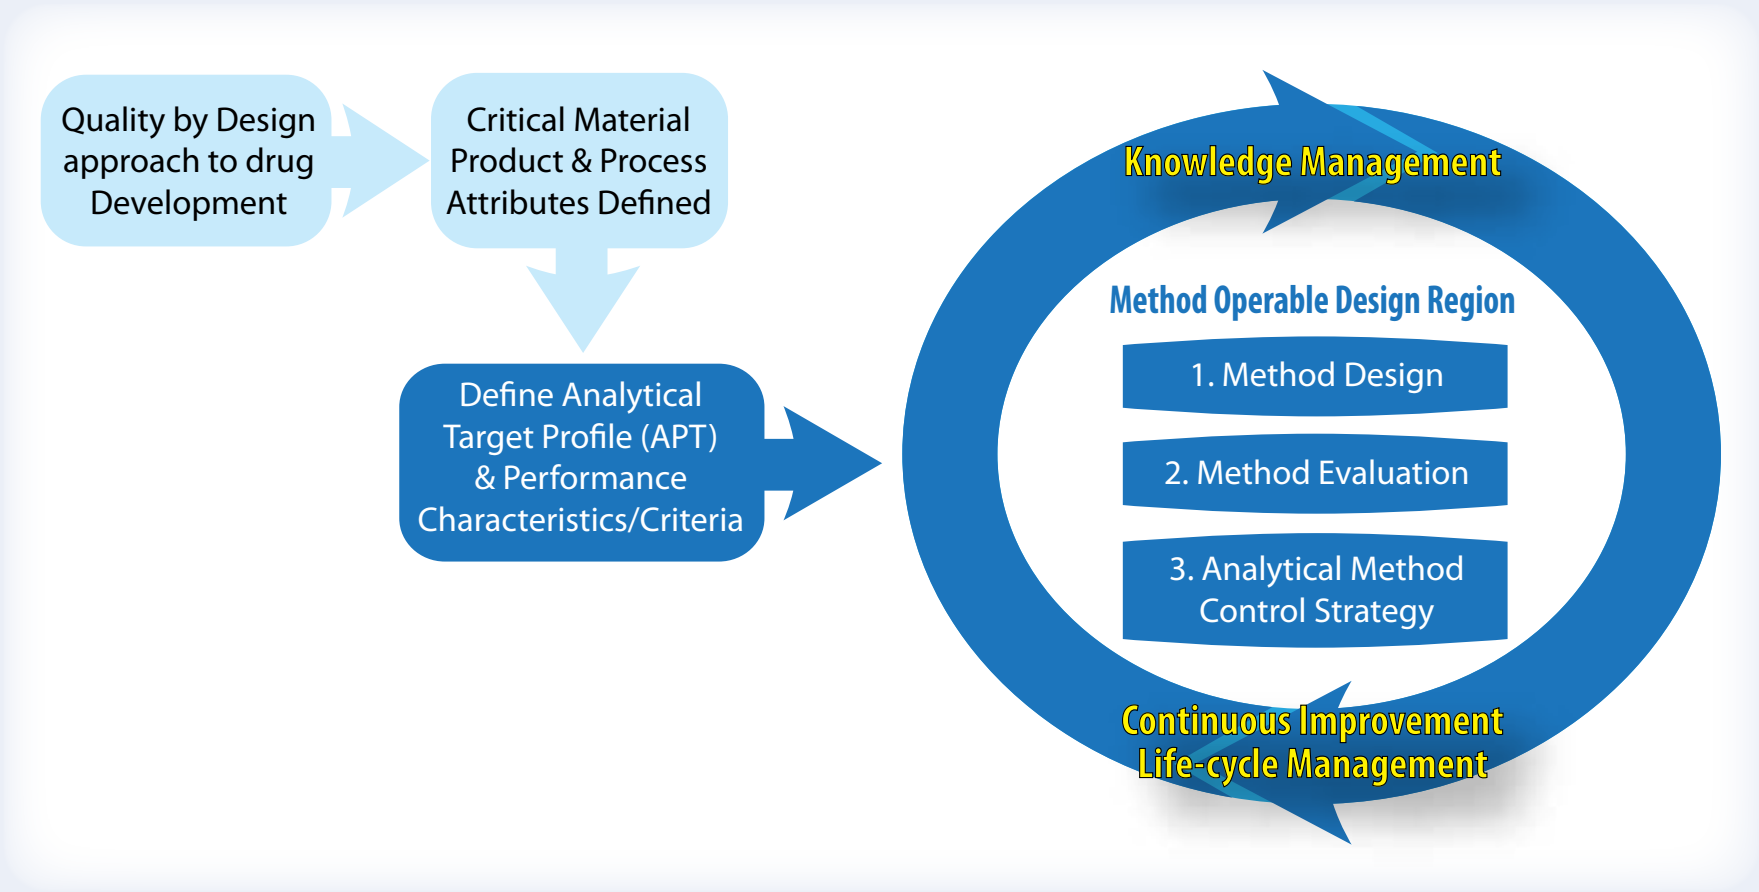
Quality by Design approach to drug Development
Critical Material Product & Process Attributes Defined
Define Analytical Target Profile (APT) & Performance Characteristics/Criteria
Knowledge Management
Method Operable Design Region
1. Method Design
2. Method Evaluation
3. Analytical Method Control Strategy
Continuous Improvement Life-cycle Management

Analytical Target Profile (ATP)—It defines what the method has to measure and to what level the measurement is required (i.e., performance level characteristics—such as precision, accuracy, working range, sensitivity—and the associated performance criteria).

Critical Quality Attributes—A quality attribute for which there is a substantial risk of impacting the safety or efficacy of a product.

Method Design—Systematic and science based method development to meet the ATP requirement.

Method Evaluation—Critical factors that may impact the method performance are evaluated with multivariate DOE or appropriate screening experiments.

Control Strategy—Define method parameter set points and operating space. Implement change control process.

Knowledge Management—All knowledge obtained from method design, evaluation and operating space definition should be retained and transferred with the method to allow future method life cycle management.

Figure 1. Components of application of quality by design (QbD) to analytical methods[1].

QbD Method Development

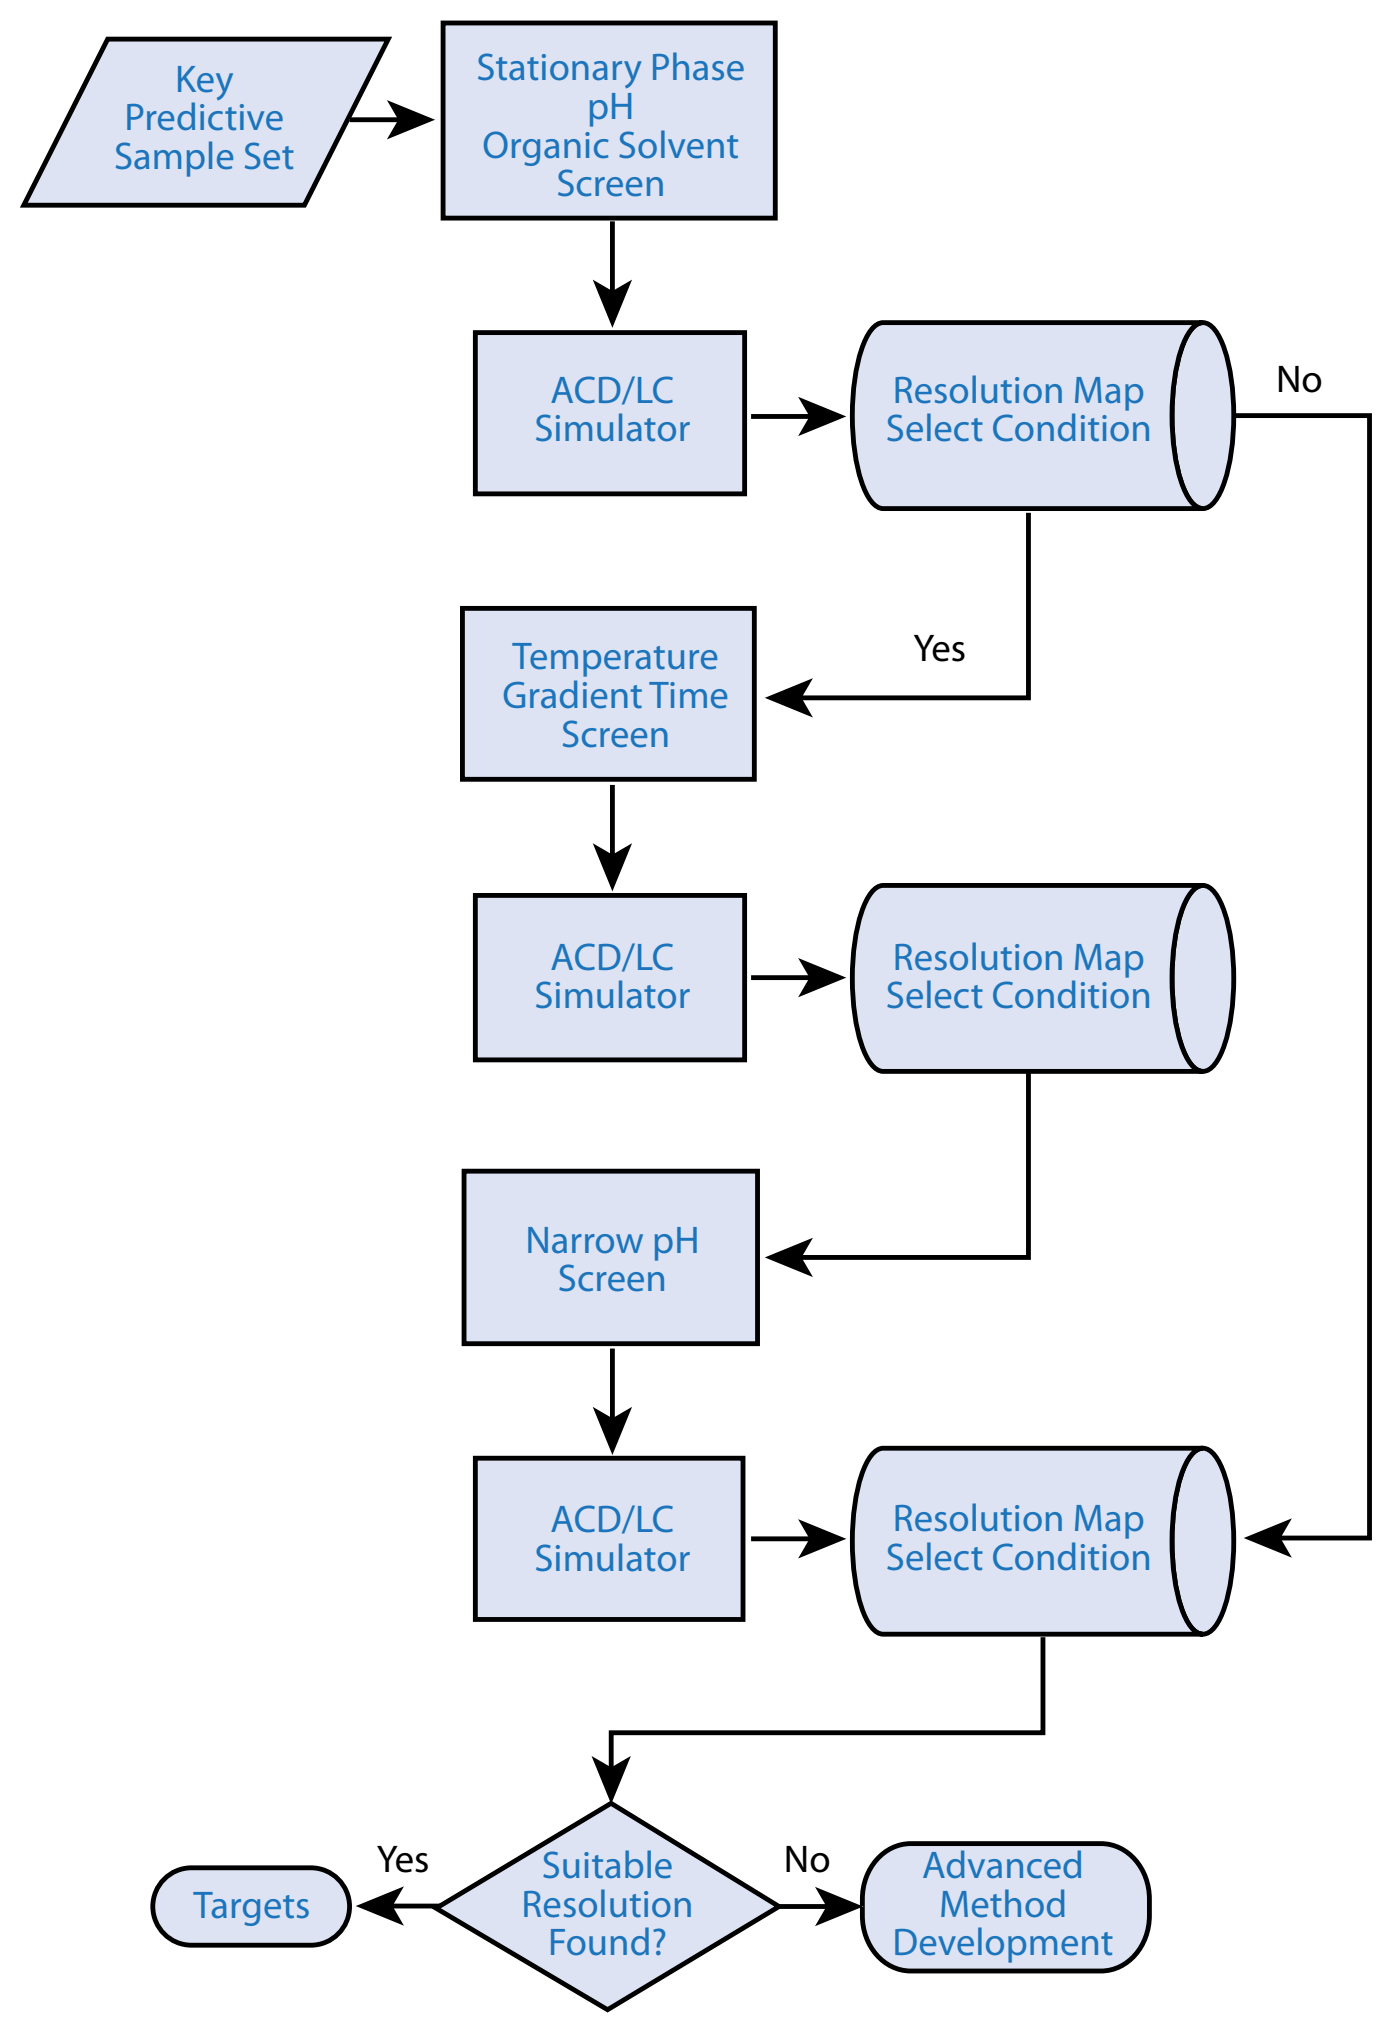
Key Predictive Sample Set
Stationary Phase pH Organic Solvent Screen
ACD/LC Simulator
Resolution Map Select Condition
No
Yes
Temperature Gradient Time Screen
ACD/LC Simulator
Resolution Map Select Condition
Narrow pH Screen
ACD/LC Simulator
Resolution Map Select Condition
Targets
Yes
Suitable Resolution Found?
No
Advanced Method Development

Figure 2. QbD RPLC Purity Method Development.

ACD AutoChrom with Empower UPLC/SQD Control

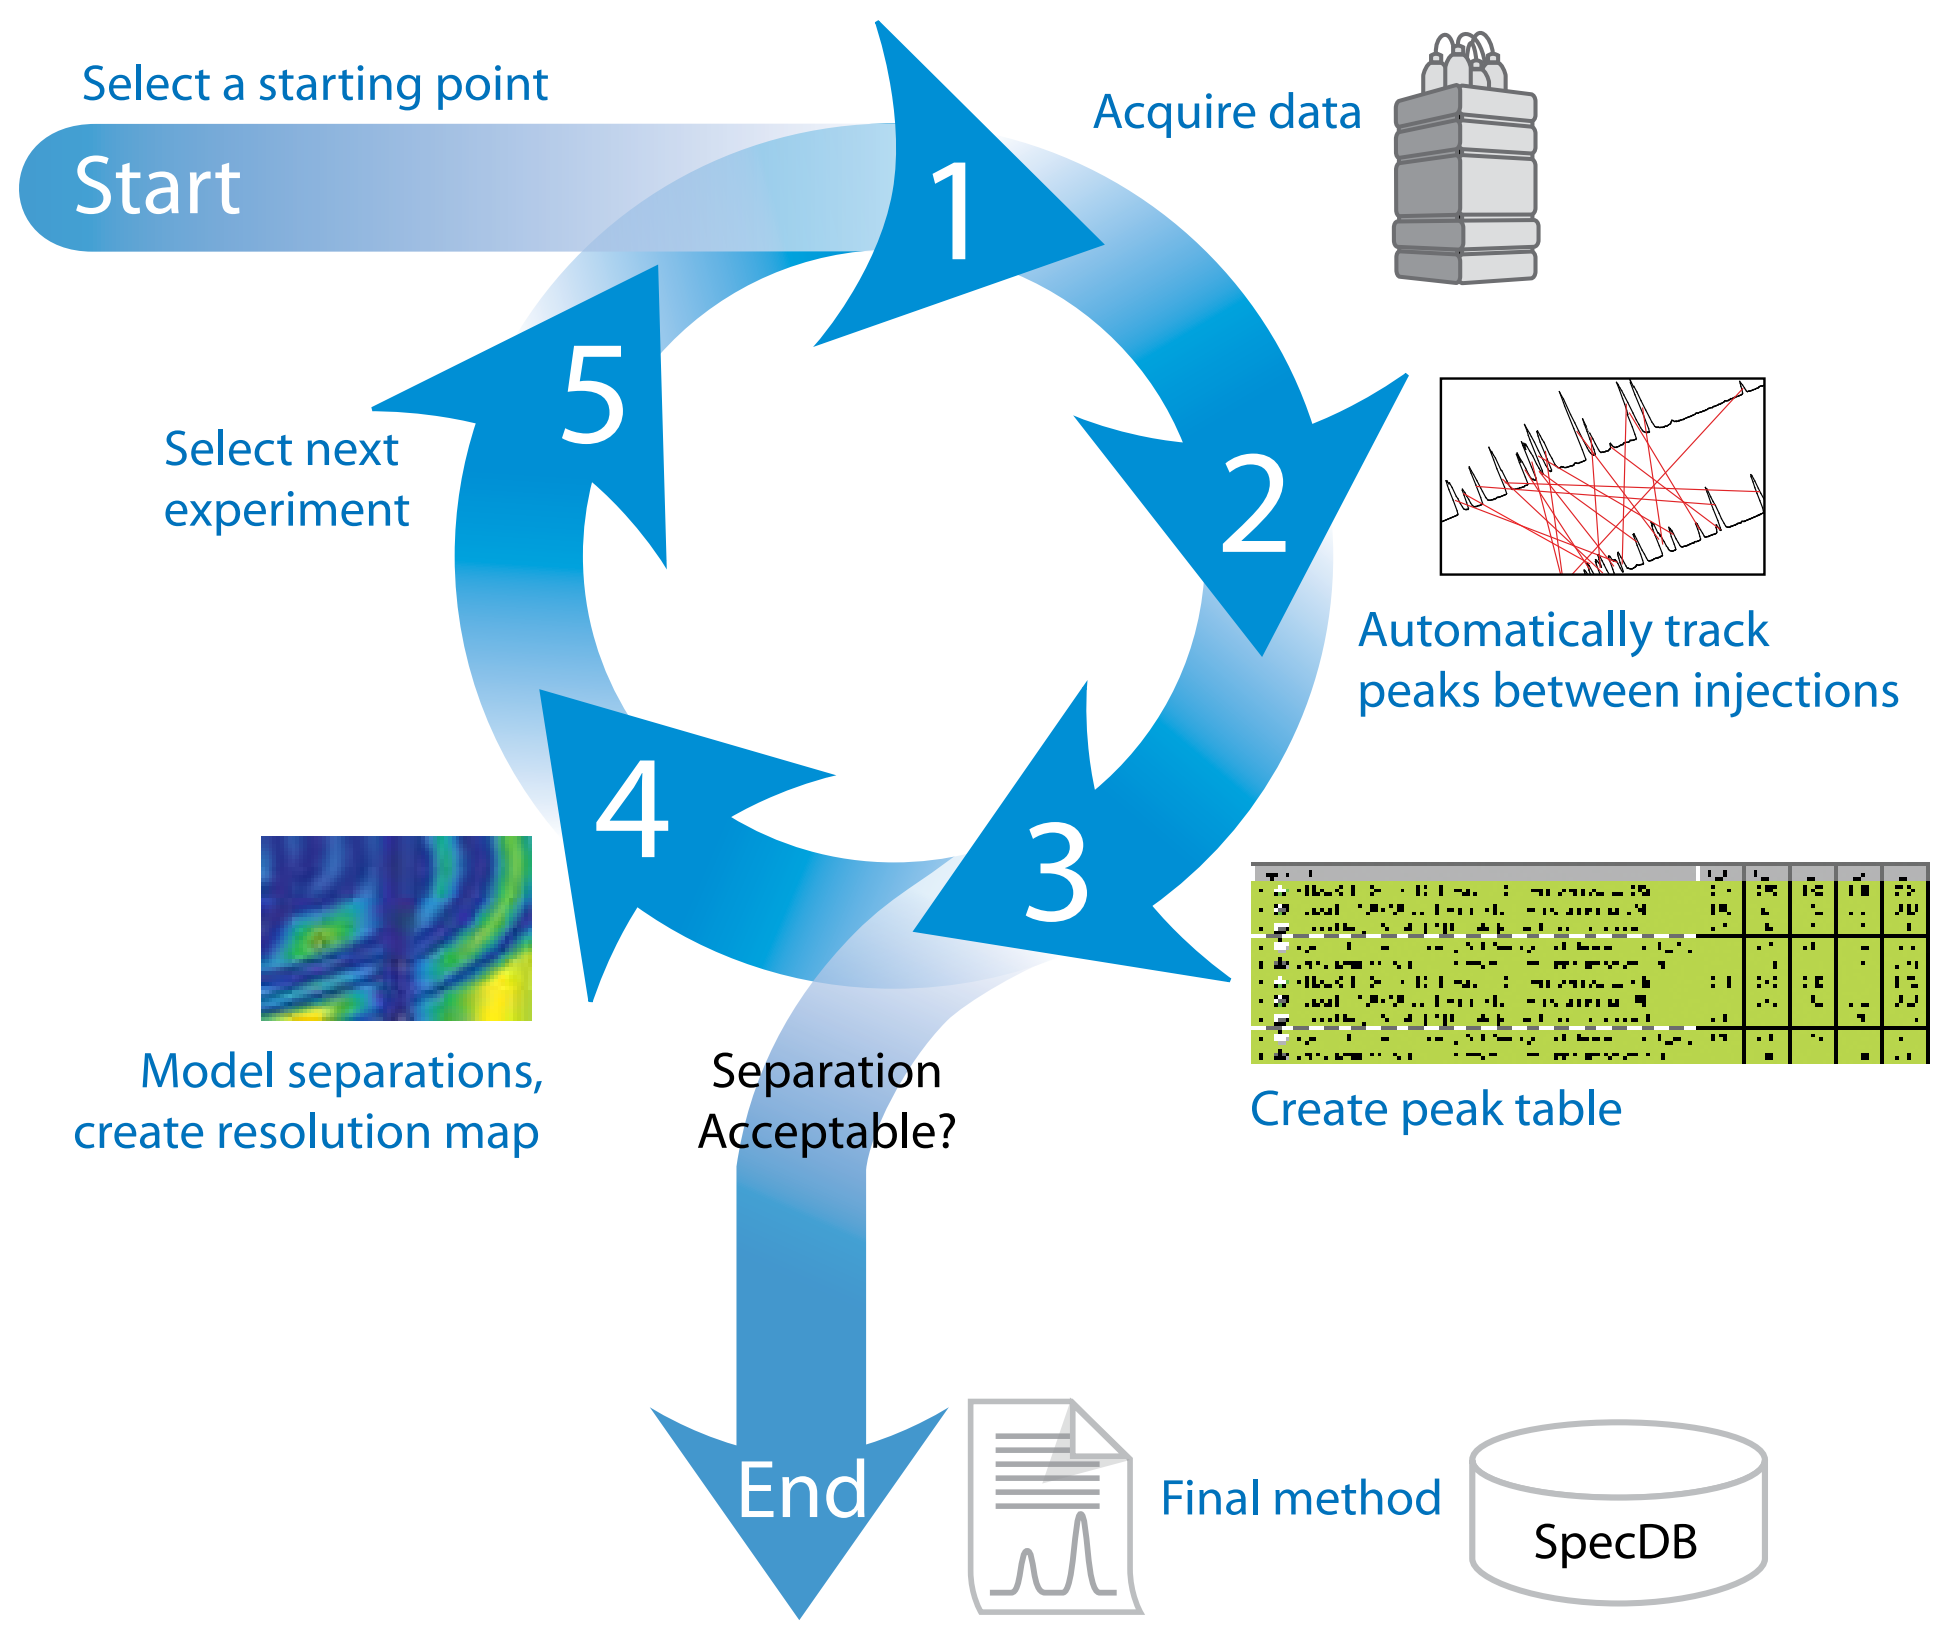
Select a starting point
Start
1
Acquire data
2
Automatically track peaks between injections
3
Create peak table
4
Model separations, create resolution map
5
Select next experiment
Separation Acceptable?
End
Final method
SpecDB

Figure 3. Automated execution of QbD RPLC method development with AutoChrom and Empower.

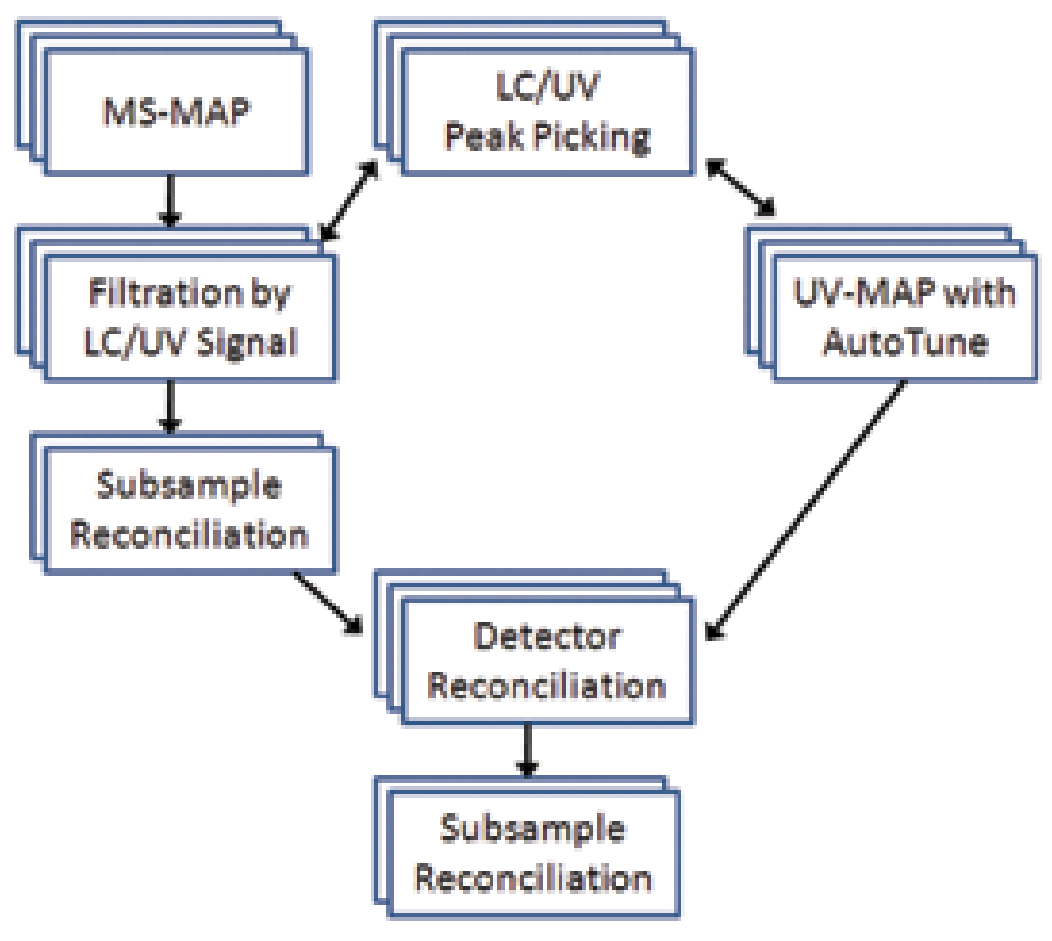
MS-MAP
LC/UV Peak Picking
Filtration by LC/UV Signal
UV-MAP with AutoTune
Subsample Reconciliation
Detector Reconciliation
Subsample Reconciliation

Figure 4. Schematics of automated data process (ADP) developed based on AutoChrom to integrate and track chromatography peaks for QbD experimentation reports.

Experimental

Automation Software: ACD/AutoChrom MDS for Empower 2 v.12.02

Structure-based Modeling: ACD/Log*D* Sol Suite v.12.02

Chromatography Data System: Waters Empower 2

Instrumentation: Waters Acquity with SQD MS detection

Results

Wave 1: Stationary phase, pH, and organic modifier screen

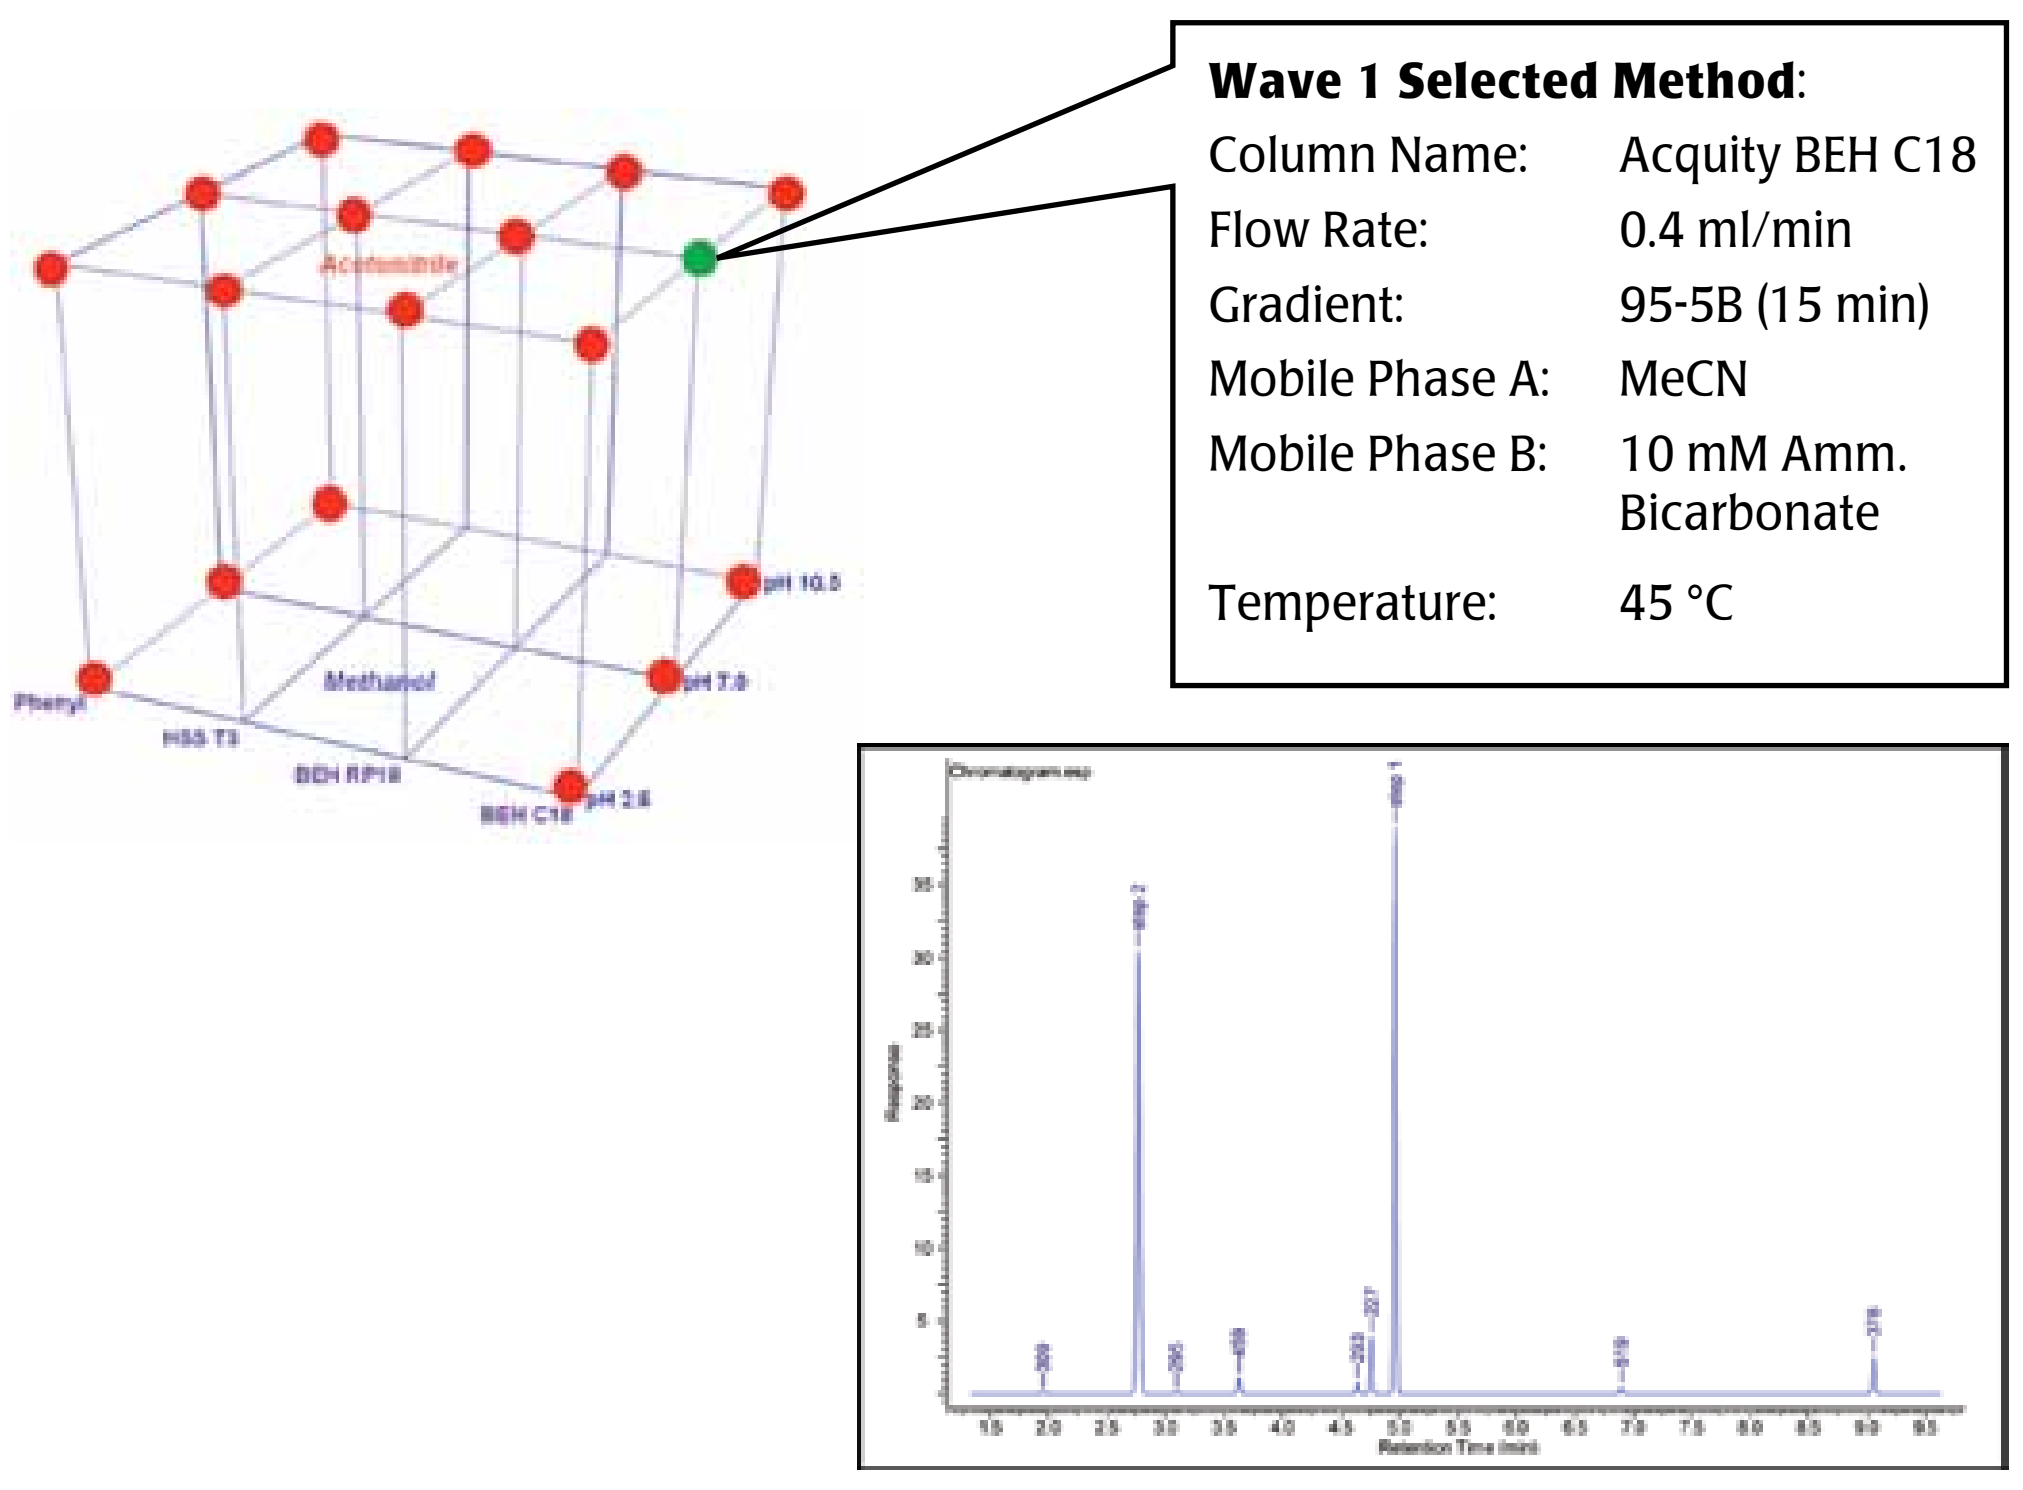
Acetonitrile
Methanol
Phenyl
HSS T3
BEH RP18
BEH C18
pH 10.5
pH 7.0
pH 2.6
Wave 1 Selected Method:
Column Name: Acquity BEH C18
Flow Rate: 0.4 ml/min
Gradient: 95-5B (15 min)
Mobile Phase A: MeCN
Mobile Phase B: 10 mM Amm. Bicarbonate
Temperature: 45 °C
Chromatogram.esp

· Some impurities failed to retain at low pH and high pH respectively

· Neutral pH showed decent selectivity and good peak shape

Wave 2: Temperature and gradient optimization

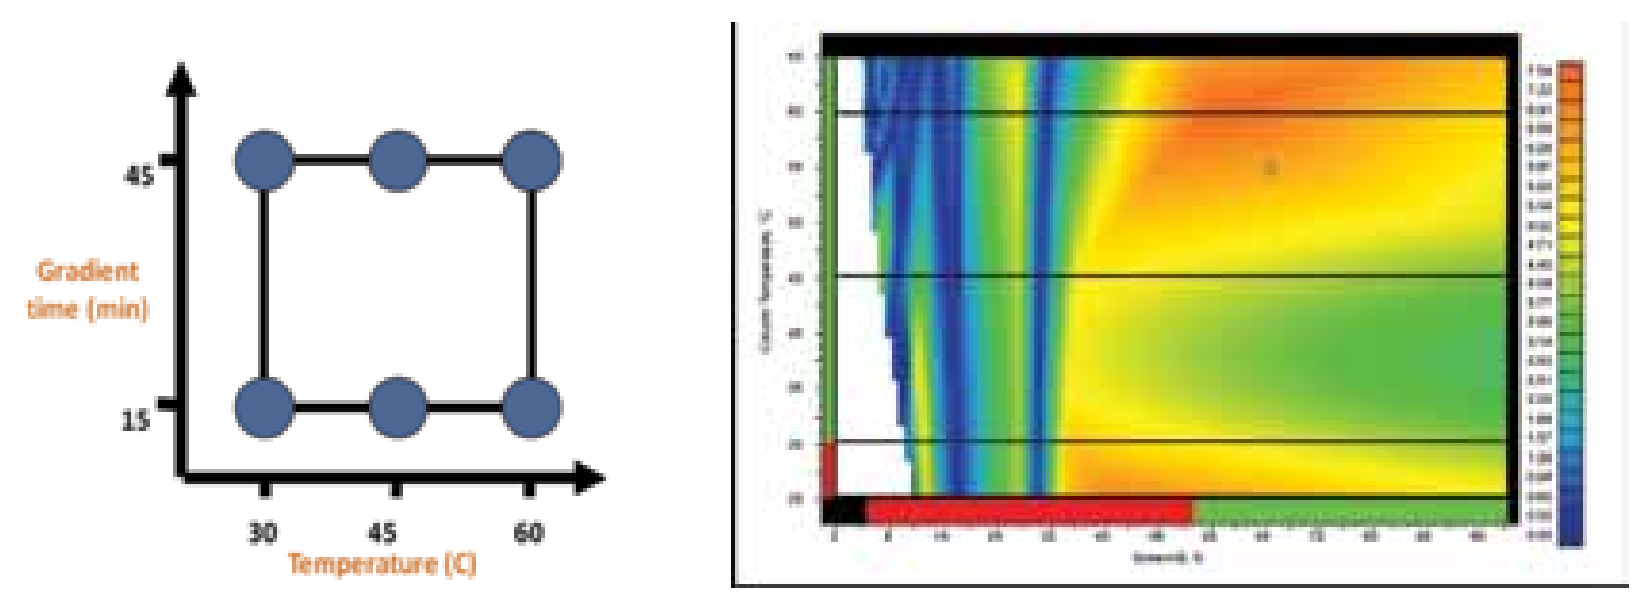
Gradient time (min)
45
15
30
45
60
Temperature (C)

Optimized Method:

BEH C18 2.1 x 100mm 1.7u, Temp=55C, Flow = .4ml/min, MP A = 10 mM Ammonium Acetate pH 6.65, MP B = MeCN, Gradient = 5% B to 65% B in 10 minutes

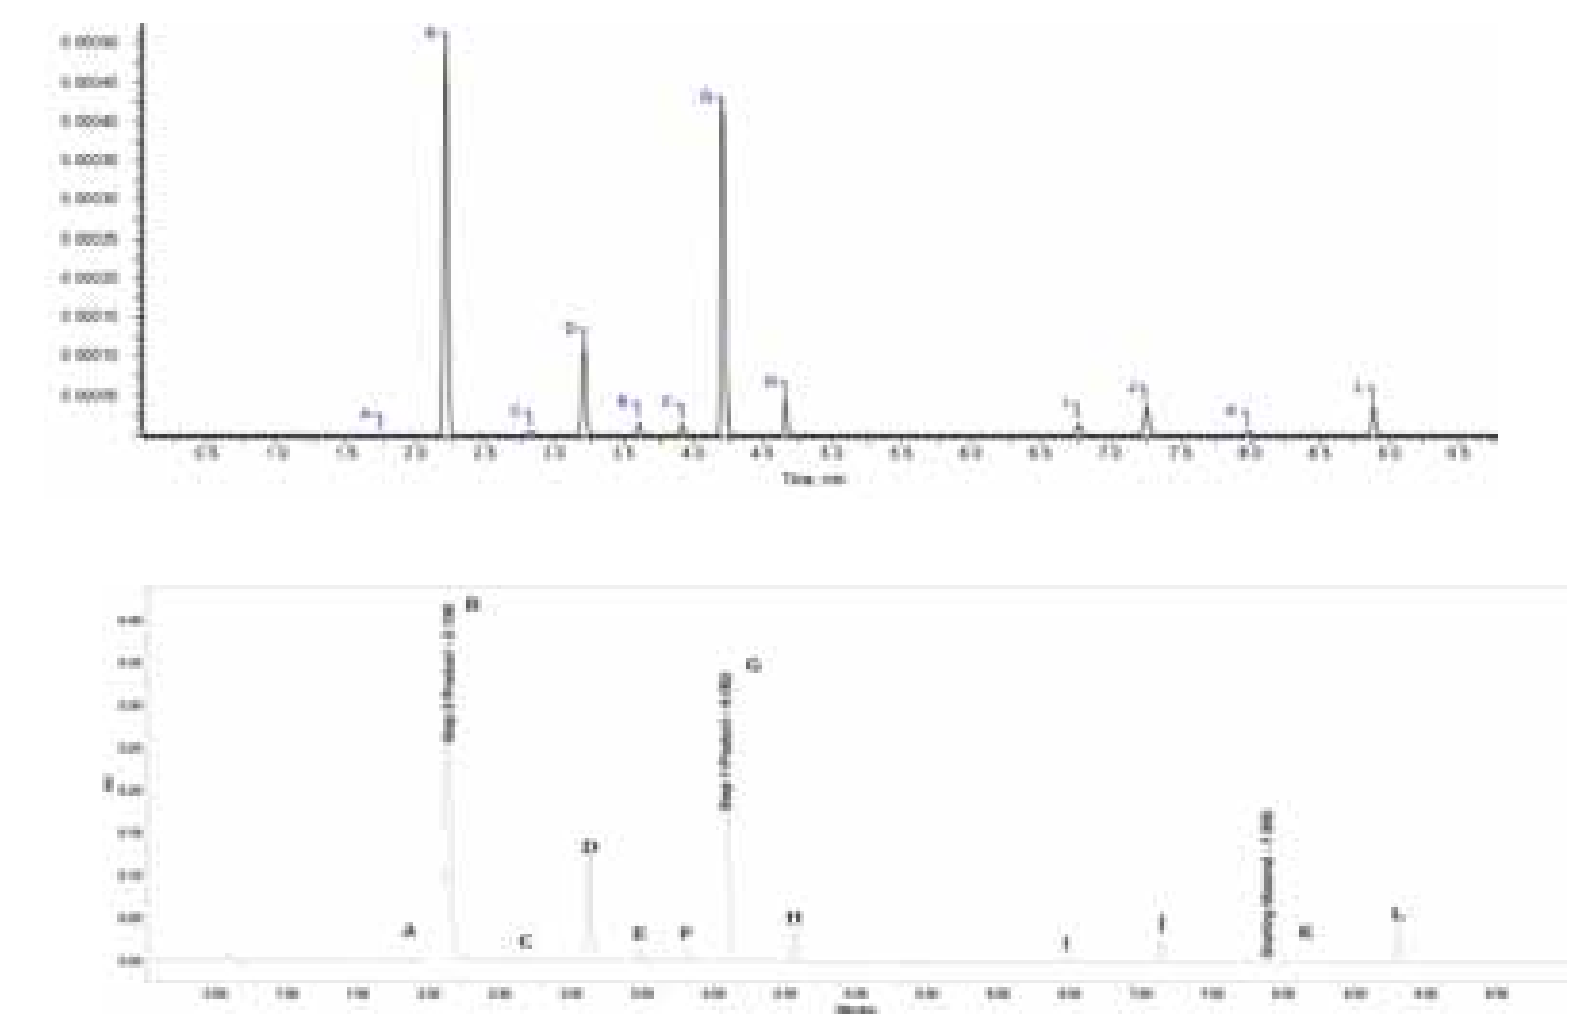

· All KPSS peaks shown. Excellent selectivity with short run time. Peak shape is excellent.

· Good correlation between simulated chromatogram and confirmation run

Advanced Chemistry Development, Inc.
Tel: (416) 368-3435
Fax: (416) 368-5596
Toll Free: 1-800-304-3988
Email: info@acdlabs.com
www.acdlabs.com

Wave 3: Narrow pH screen

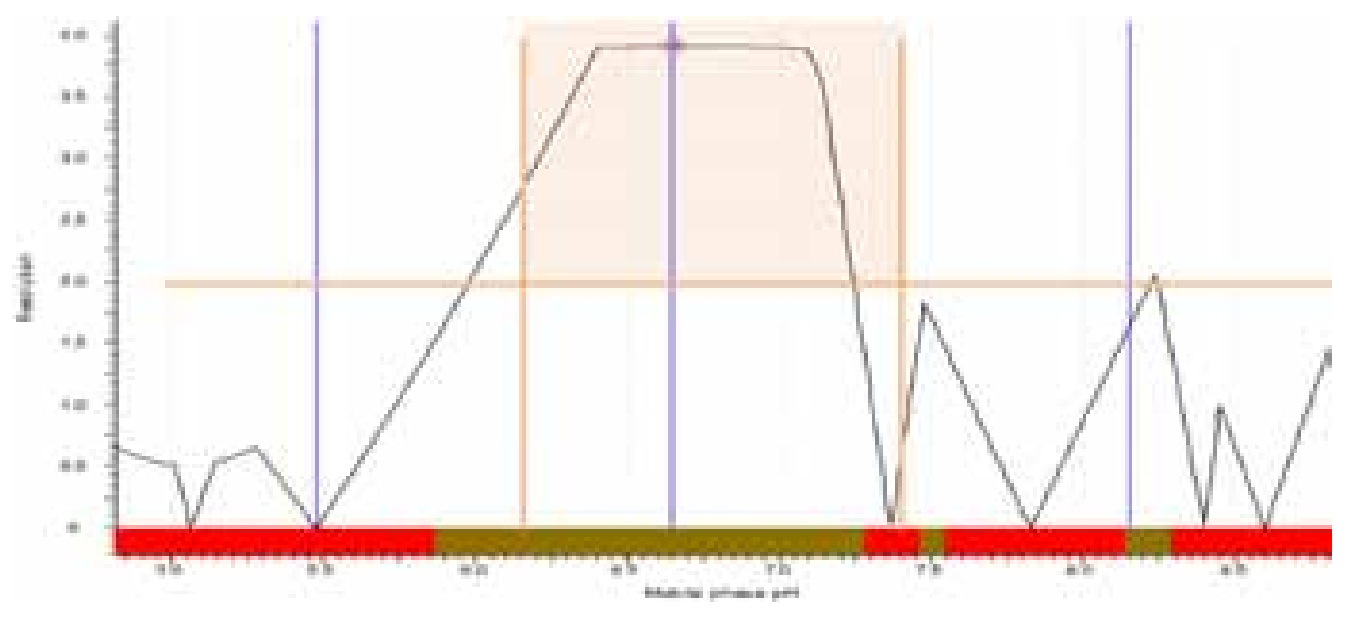

Final method:

BEH C18 2.1 x 100mm 1.7u, Temp=60C, Flow = .4ml/min, MP A = 10 mM Ammonium Acetate pH 6.65, MP B = MeCN, Gradient = 5% B to 95% B in 15 minutes

Discussions

· ACD/AutoChrom's direct control of the UPLC/SQD instrument makes the experiment setup rapid. The "intelligent" line purge and column equilibration automatically manages the required conditioning steps to help ensure data integrity.

· The Automated Data Processing (ADP) application contains all elements required to streamline the processing of the entire screening data set. Some processing parameters are being refined to minimize the required manual integration adjustment.

 · The data processing will automatically kick off once the screening completes.

 · Preset parameters will be used to integrate UV peaks and track with MS componentization.

 · A comprehensive component table will be rendered with each chemical identity tracked across all screening methods.

 · Pre-set parameters may not be ideal for all data sets, manual review and refinement is still required at the time of testing, which currently contributes to over 85% of the total analyst effort in the method development process.

· The total analyst time required for this development is 5 days.

· Following this development, risk assessment and corresponding robustness DOE was conducted to complete the definition of operating space, which is not covered here.

· Previously reported PACK application can upload all screen results to the project database for future knowledge transfer along with the final method.

Conclusions

A QbD method development system has been designed that leverages structural knowledge of the samples in question, automation, chemometric data reduction, and software-based decision support. The result is a fast, efficient system that minimizes the amount of time that the chromatographer spends with the data, but investigates a very broad scope of experimental variables. Some refinement in the data processing parameters will take place in the future as well as interfacing with the DOE statistic package to provide a complete QbD workflow from samples to operating space.

References

1. Mark Schweitzer, Matthias Pohl, Melissa Hanna-Brown, Phil Nethercote, Phil Borman, Gordon Hansen, Kevin Smith (Cephalon), Jaqueline Larew, "Implications and Opportunities of Applying QbD Principles to Analytical Measurements" *Pharmaceutical Technology*, Volume 34, Issue 2, **2010**.

2. Mike McBrien, Gang Xue, Jeff Harwood, Yanqiao Xiang, Andrey Vazhentsev, "Building a Global Chromatography Knowledgebase for Method Development Prediction" HPLC 2009, Dresden, Germany, **2009**.