Addressing the challenges of poor solubility: Rapid development and clinical evaluation of a lipid based formulation to enhance oral bioavailability of amuvatinib (MP-470)

QUOTIENT BIORESEARCH

P.D. Scholes[1], J. McDermott[1], J. Vertommen[2], J-L Colin[2], G Choy[3], M Azab[3], R Joshi[3] and S. Redkar[3]

[1]Quotient Bioresearch, Clinical Sciences, Mere Way, Ruddington Fields, Nottingham NG11 6JS, UK
[2]Capsugel, Product Development Center Strasbourg, France
[3]SuperGen Inc., Dublin CA, USA

Introduction

Physicochemical and biopharmaceutical properties of new chemical entities (NCEs) are presenting increasing challenges to successful oral drug delivery. It has been estimated that up to 70% of molecules in the industry pipeline exhibit poor solubility characteristics[1]. The typical impact of sub-optimal pharmacokinetic (PK) profiles arising from the First-in-Human (FIH) study will be cost incursions and time delays whilst an optimal formulation is identified for demonstrating Proof-of-Concept (PoC). It has been estimated that a conventional development programme can take between 12-15 months and cost more than $1.5 million to conduct[2]. A key contributor to these metrics is the convoluted CMC process arising from conventional workstreams (Figure 1). We have previously described how an integrated GMP/GCP Translational Pharmaceutics™ platform can reduce the time, cost and risk in the optimisation of drug products in early development[3].

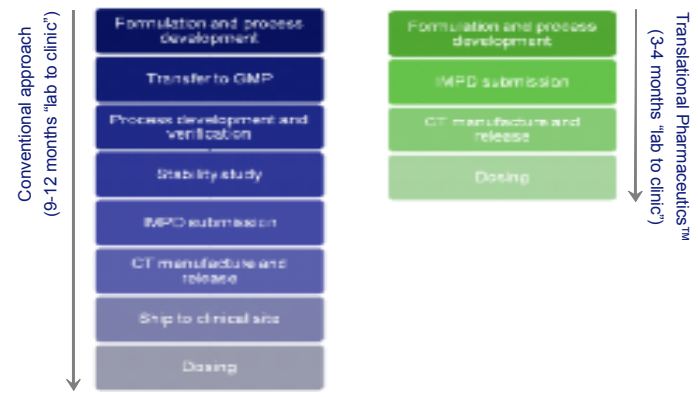

Conventional approach (9-12 months "lab to clinic")
Formulation and process development
Transfer to GMP
Process development and verification
Stability study
IMPD submission
CT manufacture and release
Ship to clinical site
Dosing
Formulation and process development
IMPD submission
CT manufacture and release
Dosing
Translational Pharmaceutics™ (3-4 months "lab to clinic")

Figure 1. Accelerating CMC development through a Translational Pharmaceutics platform

Here we present data on amuvatinib (SuperGen Inc.), a novel multi-targeted tyrosine kinase inhibitor specifically designed to be a potent inhibitor of mutant c-Kit and PDGFRα. In early clinical studies amuvatinib had exhibited solubility-limited exposure and a strong (positive) food effect. Through the use of an integrated formulation development, manufacturing and clinical evaluation programme, a lipid-based drug delivery system demonstrating significantly improved oral bioavailability was effectively identified.

Formulation Development

A suspension-type, lipid-based formulation encapsulated in a Licaps® Drug Delivery System capsule was developed for amuvatinib by Capsugel. The lead formulation containing lipid-based excipients was identified from experience-based *in vitro* screening experiments. These *in vitro* experiments included dispersion testing, digestion testing and sedimentation testing using a NIR Centrifugational Stability Analyzer (LUMiFuge®)[4]. The formulation work was complimented by the development and validation of analytical methods adapted to the lead lipid based formulation for in-process and quality control purposes. A standard Capsugel formulation characterisation programme including Differential Scanning Calorimetry (DSC) analysis, rheological measurements and density measurements was performed on the lead lipid formulation to characterise the formulation and define process parameters using a Quality by Design (QbD) approach[5].

Clinical Conduct

Clinical manufacturing and dosing were conducted within an integrated GMP/GCP Translational Pharmaceutics platform at Quotient Bioresearch. Formulation, analytical and manufacturing methodologies were transferred under protocol into a GMP environment. A "full scale" batch size of 250g was selected to cover the needs of the clinical study and quality control (QC) testing. Briefly, the formulation was prepared by melting the lipidic excipients and then adding the drug substance whilst mixing and homogenising at an elevated temperature. Formulation equivalent to 30mg amuvatinib was then filled into hard gelatin capsules. A process trial was conducted to confirm homogeneity of the lipid suspension prior to encapsulation. Representative batch release data and short-term (seven day) stability data were also generated to support the Clinical Trial Application (CTA) submission. Assay/impurities and dissolution parameters were assessed using suitably qualified reverse phase HPLC and USP <711> (Apparatus A) methods respectively.

After regulatory and ethics approvals, the amuvatinib lipid-based formulation was dosed as 3 x 30mg capsules in a two-way randomised crossover study in the fasted state in 12 healthy volunteers, along with a 100mg reference formulation (dry powder in capsule) with a 14 day wash-out period. Blood samples were taken up to 48hr post-dose for PK analysis. The relative bioavailability of the lipid-based formulation was assessed as the primary study objective.

Results

Formulation Development

A lead lipid-based suspension formulation was identified. Encapsulation process simulation data for this lead formulation are shown in Figure 2 and indicate that the formulation presents a viscosity within the recommended range of 100 to 1000 mPas when dosed at 40°C using shear rates typically generated in industrial filling pumps. After filling, the shear rate drops and the viscosity of the formulation increases on cooling of the formulation in the capsules. Further confirmation of process suitability and scaleability for downstream development was demonstrated *via* the conduct of a mixing device and addition sequence-testing study, a holding-time study and an industrial pump simulation study (data not shown). Finally, a capsule batch was successfully produced on Capsugel's liquid capsule filling and sealing (CFS) system to confirm the process feasibility.

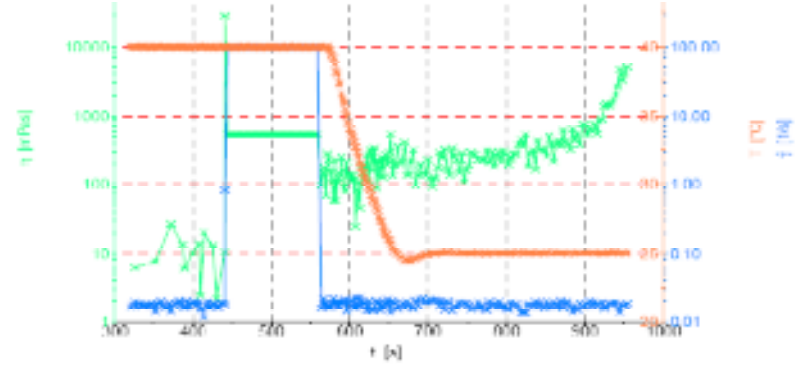

Figure 2: Encapsulation process simulation for the lead formulation

Results (continued)

GMP Technology Transfer

Representative batch release and short-term stability data were generated demonstrating the suitability of the small scale GMP manufacturing process (Table 1). Homogeneity of the product was verified through unit dose sampling at top/middle/bottom locations of the mixing vessel (Overall RSD = 1%) (data not shown). These data were successfully included in the CTA to support the "real-time" GMP manufacture and dosing of the clinical drug product.

Test	Specification	T = zero	T = 7 days	T = 28 days
Appearance	No visible residue on external capsule surface, no cracks, dents or physical defects on capsules	Complies	Complies	Complies
Assay	90.0 – 110.0 %	103.4	103.4	102.5
Related Substances	Unspecified = NMT 0.5%	Complies	Complies	Complies
	Specified = NMT 2.0%	Complies	Complies	Complies
	Total = NMT 5.0%	0.07%	0.07%	0.33%
Dissolution (USP<711> Apparatus 1	Report results	15min = 30.3% 30min = 63.5% 45 min = 96.0% 60min = 103.1%	15min = 29.7% 30min = 77.1% 45 min = 99.2% 60min = 100.8%	15min = 23.7% 30min = 72.1% 45 min = 102.5% 60min = 101.7%

Table 1. Representative batch release and stability data for lead amuvatinib lipid formulation following storage at 25°C

Clinical

Clinical data successfully demonstrated a six-fold increase in C_{max} and a two-fold increase in AUC from the amuvatinib lipid-based formulation compared to control (dry powder in capsule) (Figure 3)[6].

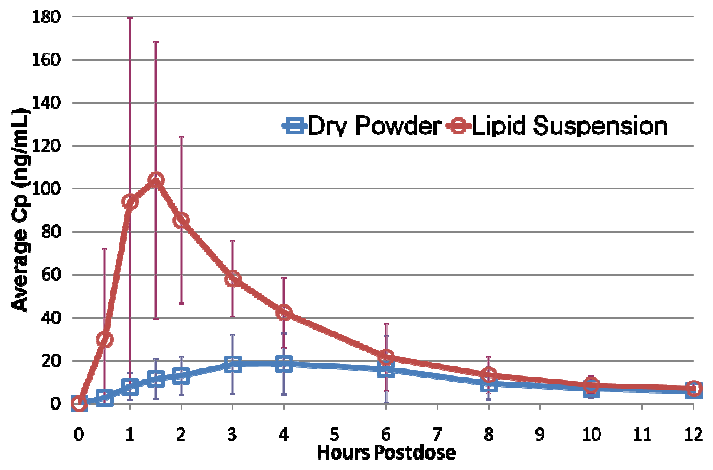

Dry Powder
Lipid Suspension
Average Cp (ng/mL)
Hours Postdose

Figure 3. Mean plasma concentration of Amuvatinib from reference and lipid formulations time.

Timelines

The application of an integrated workplan delivered significant time savings in achieving these clinical data (Figure 4). Using Capsugel's expertise a lead formulation was identified in under 12 weeks. The clinical stage including technology transfer, regulatory submissions, clinical manufacturing and dosing was subsequently conducted in under 14 weeks at Quotient Clinical.

Conclusions

Early development strategies for the formulation optimisation of drug products with specific drug delivery needs are enhanced *via* the integration of pharmaceutical and clinical processes and facilities. Such approaches can enhance the speed and outcome for the development of enabled formulations to achieve satisfactory exposure or PK profiles in FIH and Phase I studies.

The development and use of a lipid-based formulation for amuvatinib, a BCS Class II compound, encapsulated in a Licaps® Drug Delivery System, has been shown to improve oral bioavailability significantly in comparison to a reference dry powder in capsule formulation. In addition significant time savings in the development process have been realised through the utilisation of a combined GMP/GCP platform (Figure 4).

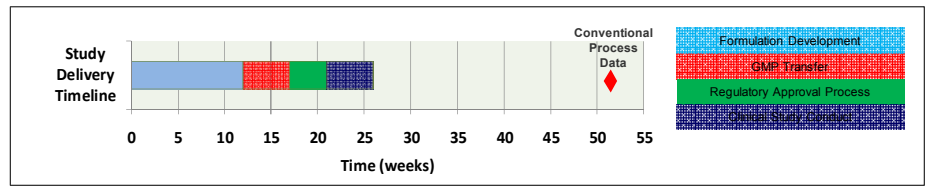

Study Delivery Timeline
Conventional Process Data
Formulation Development
GMP Transfer
Regulatory Approval Process
Time (weeks)

Figure 4. Overall study timelines for pharmaceutical development and clinical assessment of a lipid-based amuvatinib formulation.

The ability to manufacture product in a GMP environment close to the point of clinical dosing reduces the stability data package required to support an extended shelf-life, and can therefore enable both an expedited regulatory submission and study conduct. This is complimented by the ability to reduce the QC testing burden through the use of appropriate in-process controls and the risk-based justification not to perform certain time-consuming finished product characterisation, such as microbiological assessments. First-subject-first-dose can be achieved within five days of the onset of the GMP manufacture of the dosage form.

References

1 Hauss (2007), Advanced Drug Delivery Reviews, 59, 667-676
2 Quotient Bioresearch market research - unpublished
3 Scholes et al (2009), AAPS Poster #2869
4 Igonin at al (2009), AAPS, Poster #3329
5 Igonin et al (2007), AAPS Poster #3356
6 Joshi et al (2009), AACR Abstract #B209

CAPSUGEL

SuperGen

Tel: +44 (0) 1638 720500 Email: info@quotientbioresearch.com www.quotientbioresearch.com